7148

Small molecule inhibitor of FEN1 nuclease utilizing a novel metal-binding pharmacophore synergizes with inhibitors of USP1, PARP, PARG and ATR

Jason Munguia, Sanjay Agarwalla, Elaine Guo, Dave Martin, Junhua Fan, Dave Lonergan, David Puerta, Zachary Zimmerman, Konstantin Taganov
Blacksmith Medicines, San Diego, CA

Abstract

Flap endonuclease 1 (FEN1) is a structure-specific metallonuclease that cleaves 5' DNA flaps during replication and repair. FEN1 is an attractive target for development of anticancer therapeutics because it is overexpressed in many tumor types and has a large number of synthetic lethality partners including genes in Homologous Recombination (HR) pathway (Mengwasser et al., 2019; Guo et al., 2020).

Utilizing fragment-based drug discovery (FBDD) approach we identified a novel metal-binding pharmacophore that binds to the two magnesium ions in the FEN1 active site. Further elaboration using fragment growth strategies resulted in highly potent and selective inhibitors. The current lead (BSM-1516) is ~65-fold more potent against FEN1 than its related enzyme Exonuclease 1 (Exo1) in biochemical assays (IC50 of 7 nM and 460 nM, respectively), an improvement of more than an order of magnitude in selectivity compared to earlier efforts. FEN1 target engagement in live cells was validated by cellular thermal shift assay (CETSA™ EC50 of 24 nM). Inhibition of FEN1 led to its increased association with chromatin in S-phase cells and recruitment of PARP1 enzyme.

In clonogenic assay, BRCA2-deficient DLD1 cells were ~15-fold more sensitive to FEN1 inhibition than their isogenic BRCA2-wild-type counterparts (EC50 of 350 nM and 5 µM, respectively), confirming the increased susceptibility of HR deficient cancer cells to FEN1 inhibition. Treatment of BRCA2-deficient but not wild-type DLD1 cells with BSM-1516 resulted in cell cycle arrest and dose-dependent decrease of S-phase BrdU incorporation into DNA. Cell cycle arrest of BRCA2-deficient DLD1 cells was accompanied by DNA damage signaling and by accumulation of chromatin-bound RPA32, a marker of ssDNA.

To explore synthetic lethal interactions of FEN1 with other DNA repair genes we performed FEN1-inhibitor-anchored CRISPR screen. This analysis revealed that in addition to HR pathway inactivation, genomic perturbations in EXO1, USP1 and PARP1 genes sensitized cells to FEN1 inhibition. Synergistic relationships of BSM-1516 and its combination potential were further explored in viability studies with a panel of DDR inhibitors (n=25) in BRCA2-proficient and deficient cell lines. Strong synergy was identified with multiple drug classes that included inhibitors of USP1 (KSQ-4279), PARP (Olaparib, Niraparib, Talazoparib, AZD5305), PARG (PDD 00017273) and ATR (AZD6738, VE-822, Elimusertib).

In vitro ADME assays and in vivo PK studies showed that BSM-1516 has properties suitable for in vivo testing, either as a single agent or in combination with synergistic DDR inhibitors, an investigation that is currently underway.

Biochemical assays: Improved potency and greater selectivity of BSM-1516 compared to historic FEN1 inhibitors

Company	Structure	ID	FEN1 IC_{50} (µM)	EXO1 IC_{50} (µM)	FEN1 vs. EXO1 selectivity (fold)
Astra Zeneca	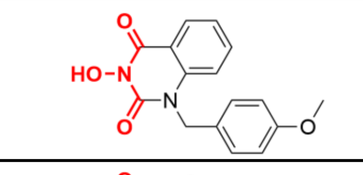	Cmpd 4	0.21	0.72	3.4
Athersys	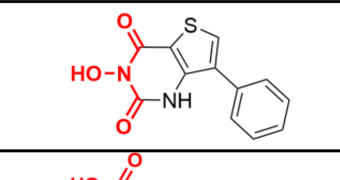	Cmpd 8*	0.13	0.41	3.2
Ideaya	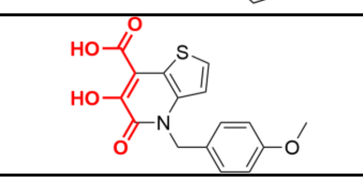	Cmpd 12	2.7	14	5.2
Blacksmith Medicines	Compound utilizing a novel metal-binding pharmacophore	BSM-1516	0.007	0.46	65

*Compound 8 cell activity and in vivo efficacy have been reported in Guo et al., PNAS 2020

BSM-1516 is novel small molecule inhibitor of FEN1 discovered using a proprietary FBDD approach that involves screening of the metal-binding pharmacophore library followed by rounds of elaboration and computational modeling.

Potency of BSM-1516 and reference compounds against FEN1 and EXO1 (members of RAD2 family of structure-specific metallonucleases) was measured in enzymatic assays. These fluorescence-based assays were adapted from van Pel et al., PLOS Genetics 2013 and utilized a flap-containing synthetic substrate carrying a fluorophore on the 5' flap and a quencher at the opposite end of the DNA oligonucleotide.

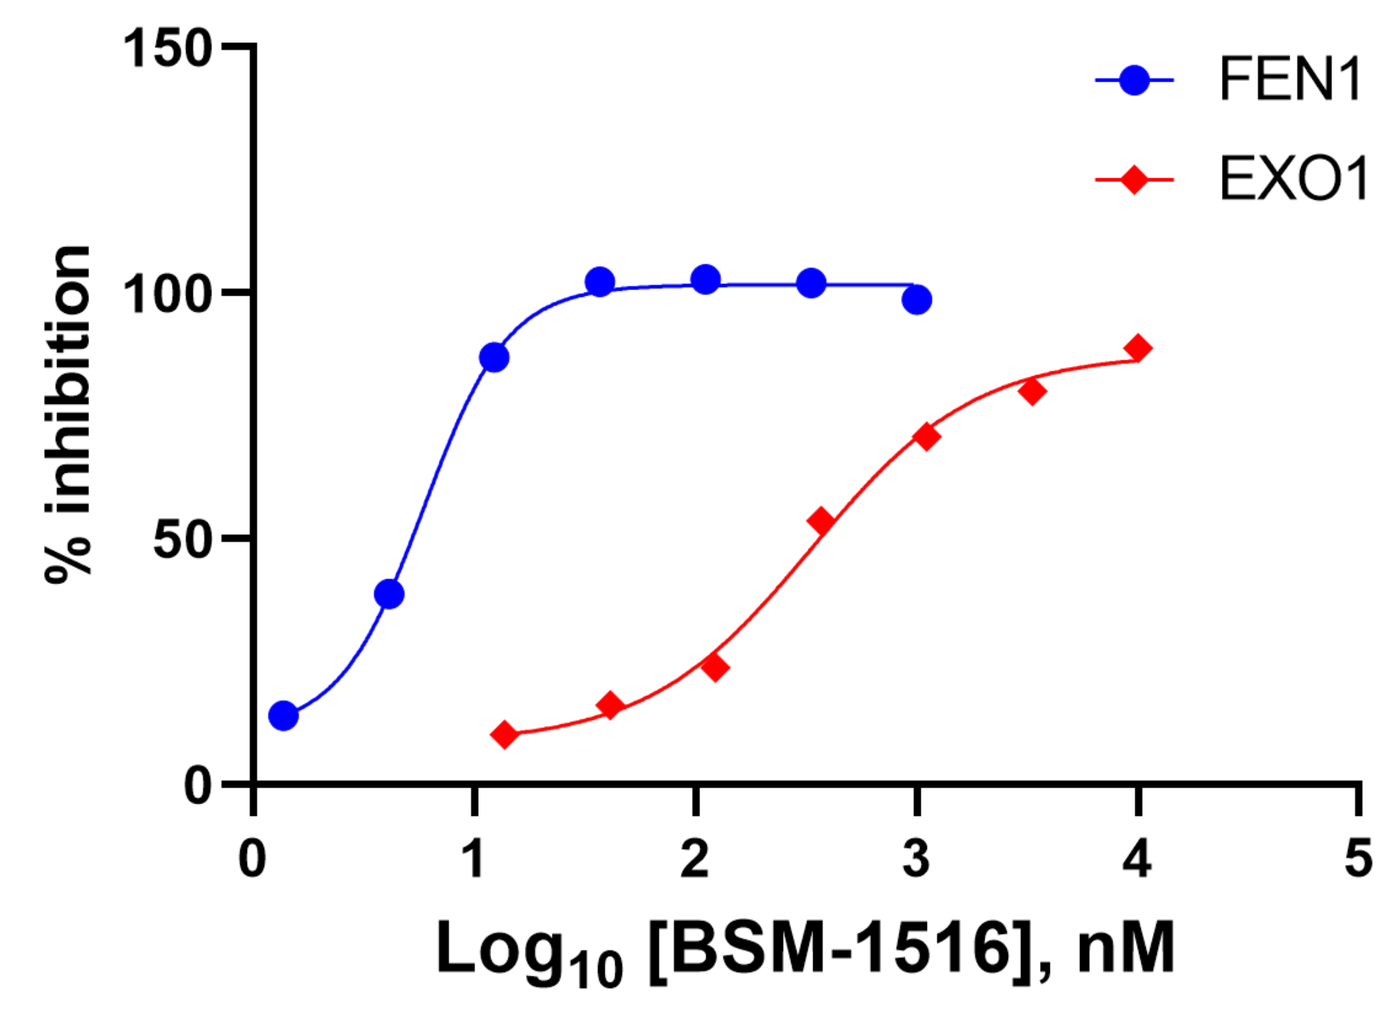
FEN1
EXO1
% inhibition
Log10 [BSM-1516], nM

Cellular Target Engagement and Mechanism of Action

BSM-1516 stabilizes FEN1 and increases its chromatin retention in S phase that is accompanied by slowdown of DNA replication and Talazoparib-mediated PARP1 trapping.

Company	ID	CETSA EC_{50} (µM)
Astra Zeneca	Cmpd 4	1.2
Athersys	Cmpd 8	0.21
Ideaya	Cmpd 12	>100
Blacksmith Medicines	BSM-1516	0.024

Cellular thermal shift assay (CETSA™) was performed using InCELL Pulse™ target engagement assay (DiscoverX) in stably transfected HEK293 cells expressing FEN1 catalytic domain with C-terminal ePL tag.

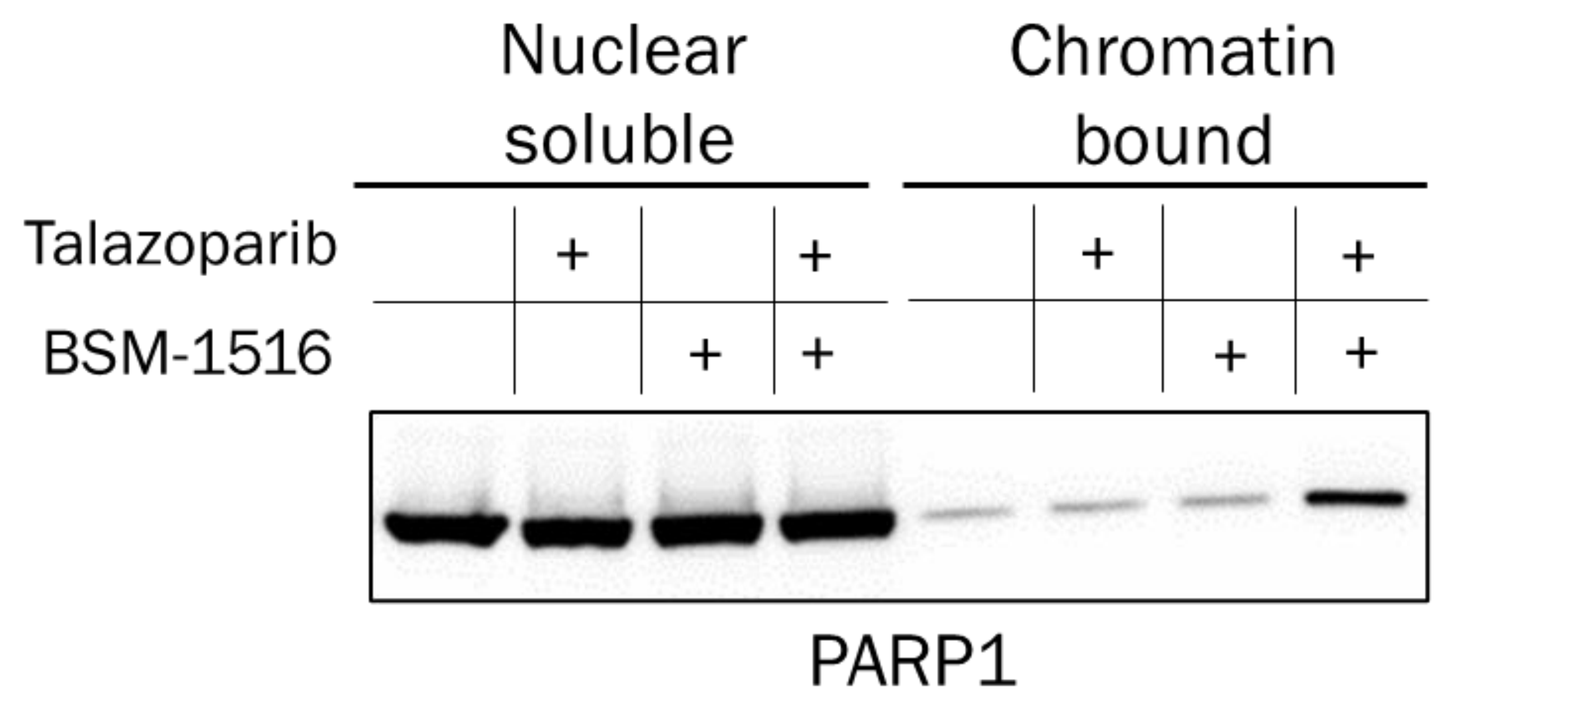
Nuclear soluble
Chromatin bound
Talazoparib
BSM-1516
PARP1

Western blot analysis of endogenous chromatin-bound PARP1 in DLD1 cells treated with indicated inhibitors for 3h and then fractionated.

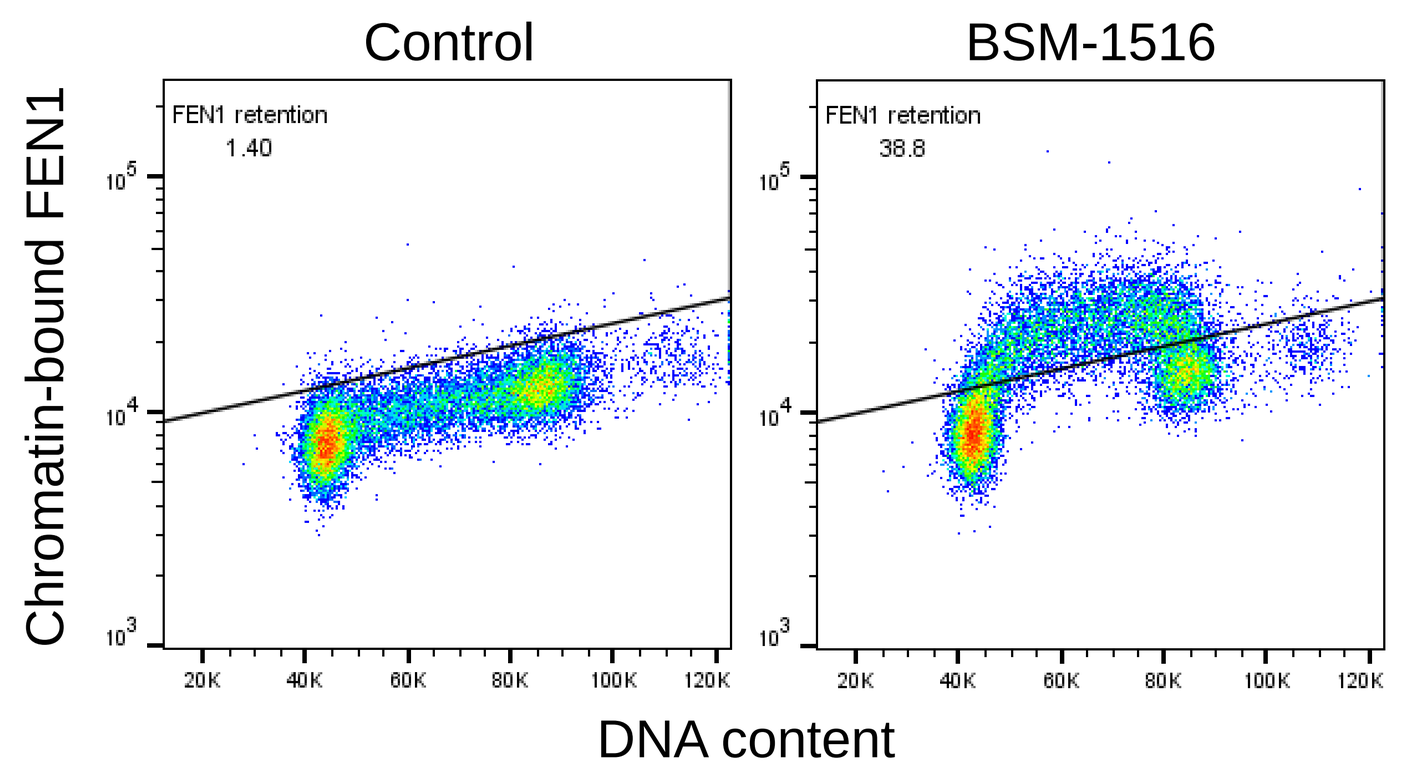
Control
BSM-1516
FEN1 retention 1.40
FEN1 retention 38.8
Chromatin-bound FEN1
DNA content

Intracellular staining of endogenous chromatin-bound FEN1 in DLD1 cells treated with 3µM BSM-1516 for 16h. Soluble FEN1 was extracted with detergent prior to fixation and subsequent analysis by flow cytometry.

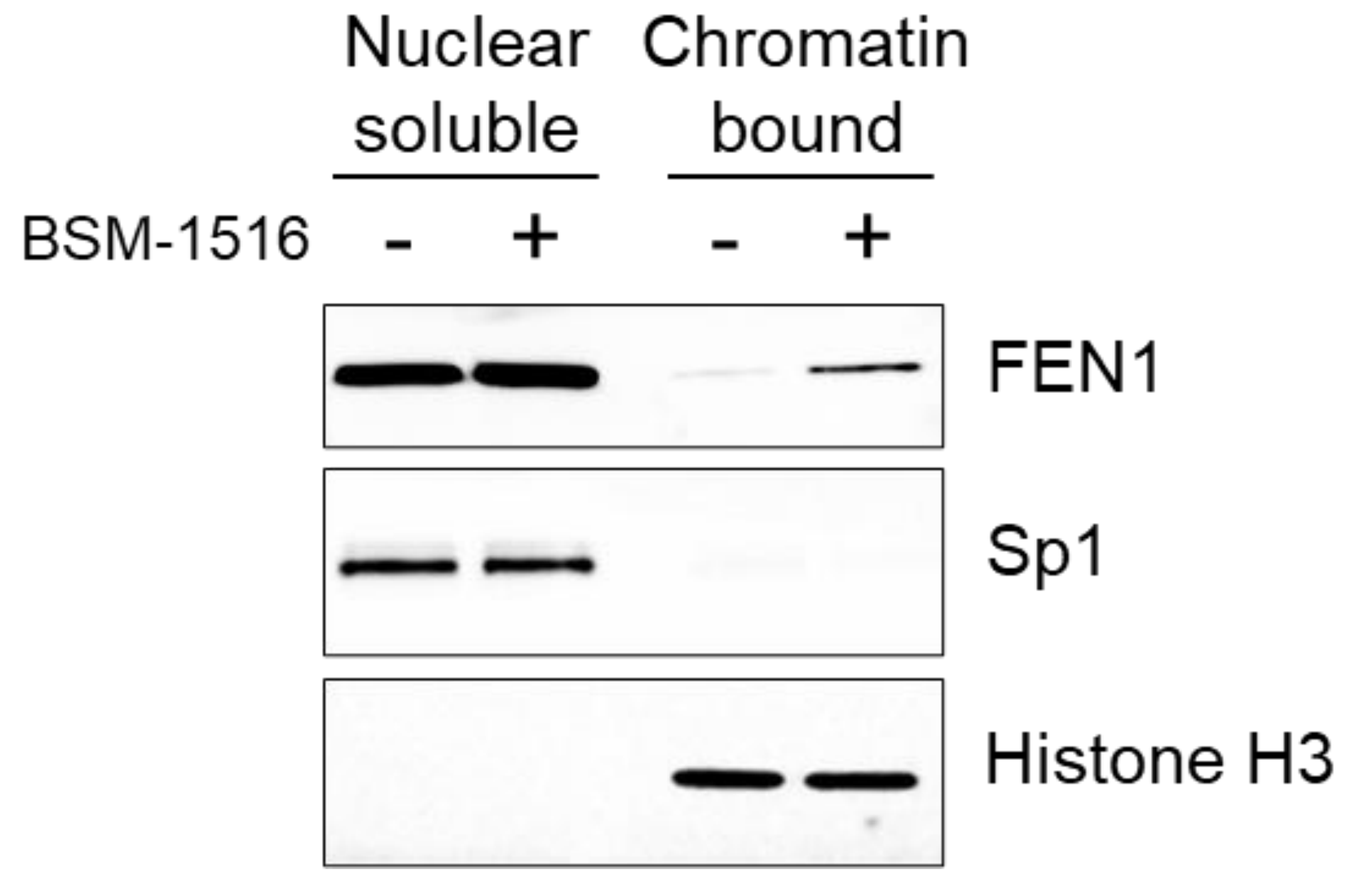
Nuclear soluble
Chromatin bound
BSM-1516
FEN1
Sp1
Histone H3

Western blot analysis of endogenous chromatin-bound FEN1 in S phase. DLD1 cells were arrested by double thymidine block, released for 3h into fresh media with or without 3µM BSM-1516 and then fractionated.

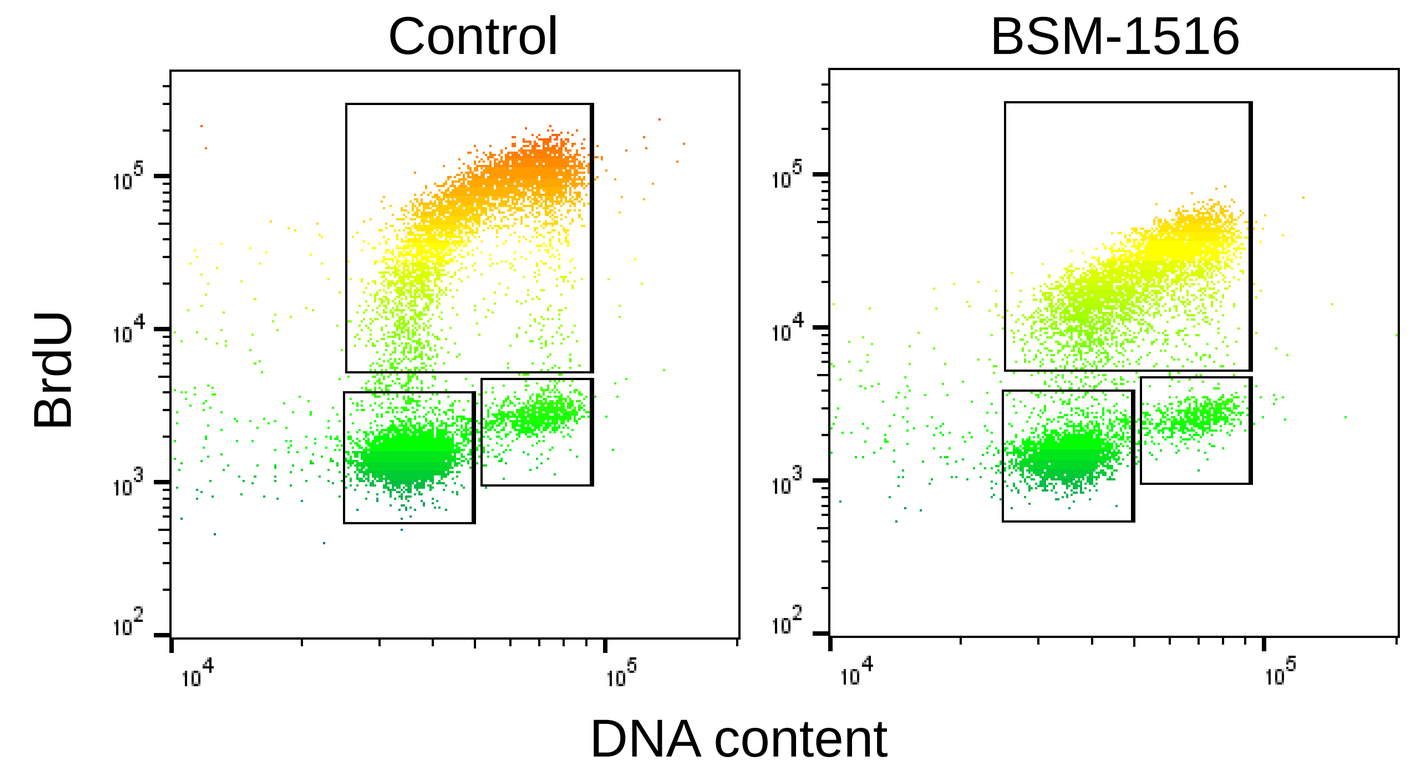
Control
BSM-1516
BrdU
DNA content

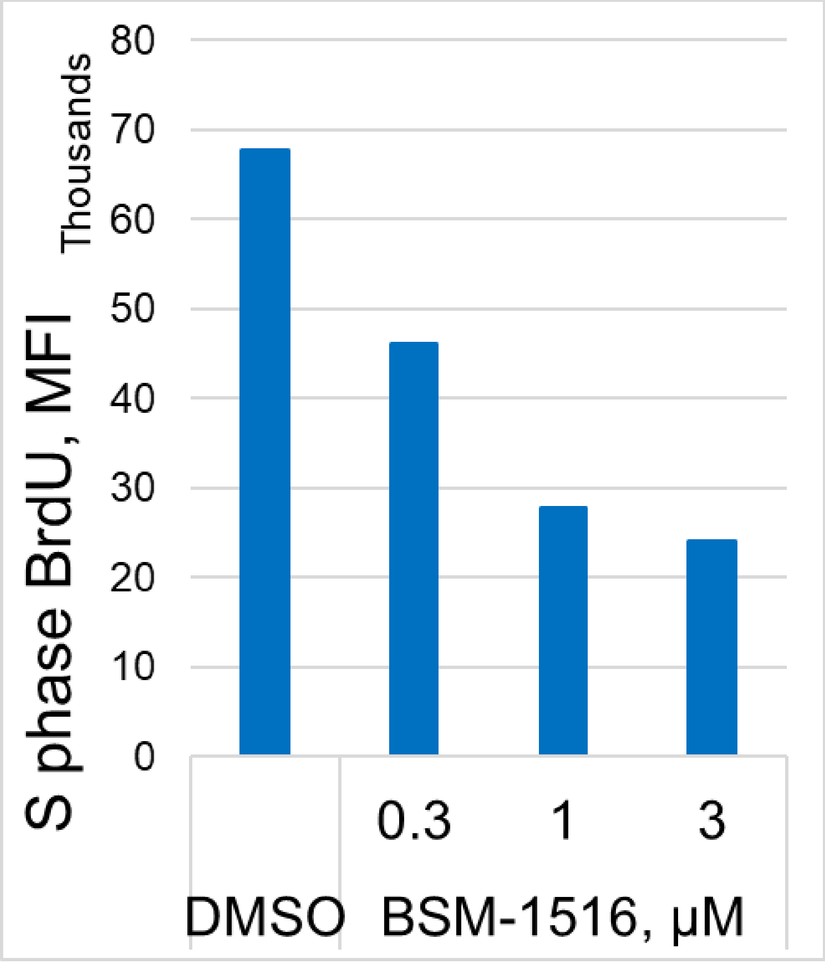
S phase BrdU, MFI
Thousands
DMSO
0.3
1
3
BSM-1516, µM

BSM-1516 causes replication suppression and impaired S phase progression. Analysis of BrdU incorporation and total DNA content monitored by flow-cytometry after 16h of BSM-1516 treatment.

CRISPR screen identifies genes that are synthetic lethal with FEN1 inhibition

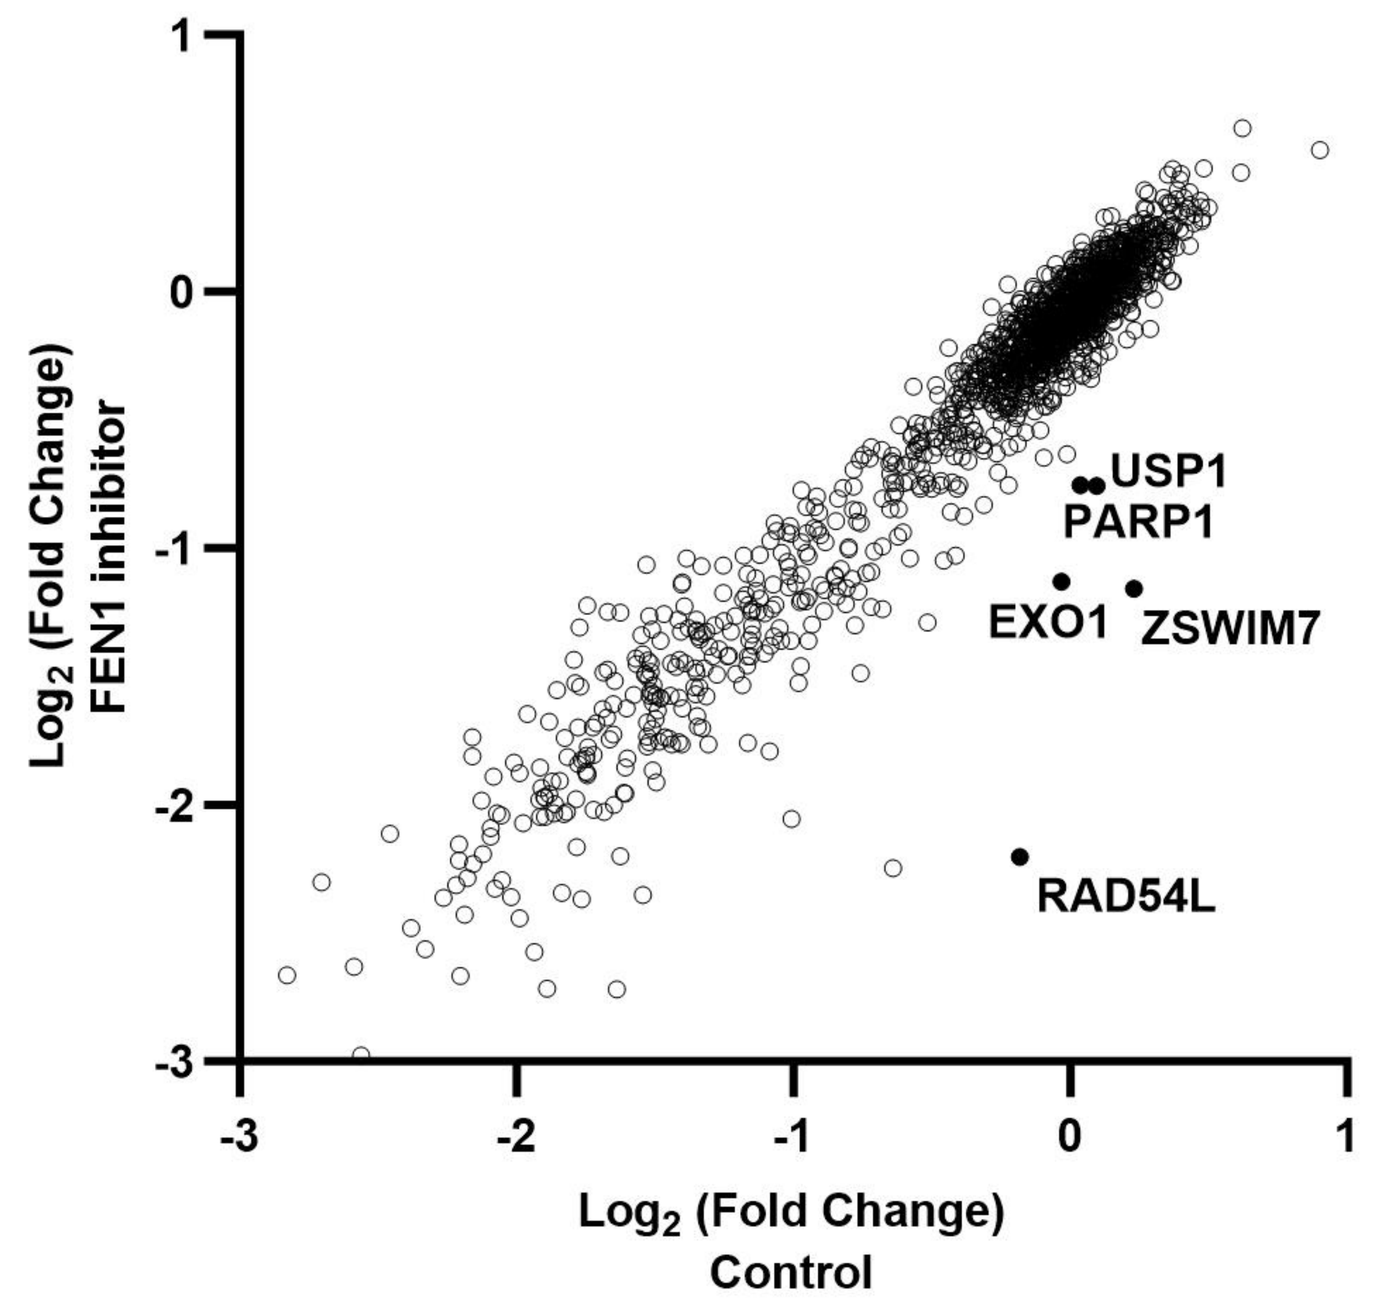
USP1
PARP1
EXO1
ZSWIM7
RAD54L
Log2 (Fold Change) FEN1 inhibitor
Log2 (Fold Change) Control

To identify genetic determinants driving FEN1 inhibitor sensitivity we performed CRISPR-Cas9 loss-of-function screen using lentiviral sgRNA sublibrary that targets DNA damage response genes and metalloenzymes. Data on the graph were plotted as ratio of sgRNA abundance on day 14 versus day 0 for control and for FEN1-inhibitor (2µM) treated DLD1 cells.

Our screen confirmed several previously published FEN1 gene interactors including USP1 (Fielden et al., 2023 bioRxiv), PARP1 (Hanzlikova et al., 2018), EXO1 (Tishkofff et al., 1997) and homologous recombination repair (HRR) pathway defects (Mengwasser et al., 2019; Guo et al., 2020). PARG and WDR48 genes were not targeted by the library.

Viability assays: BRCA2-deficient cells are >10 fold more sensitive to FEN1 inhibitor than isogenic wild-type control

Consistent with the previous reports (Mengwasser et al., 2019; Guo et al., 2020) BRCA2 gene deficiency causes sensitivity to pharmacologic inhibition of FEN1. In cell survival and culture confluency assays DLD1 cells carrying a truncating mutation of BRCA2 gene were more than 10-fold more sensitive to FEN1 inhibition than BRCA2-proficient isogenic control cells. Additionally, BRCA2-deficient cells show higher levels of RPA-coated ssDNA in G2, RPA2 S33 phosphorylation and G2/M arrest in response to BSM-1516 treatment.

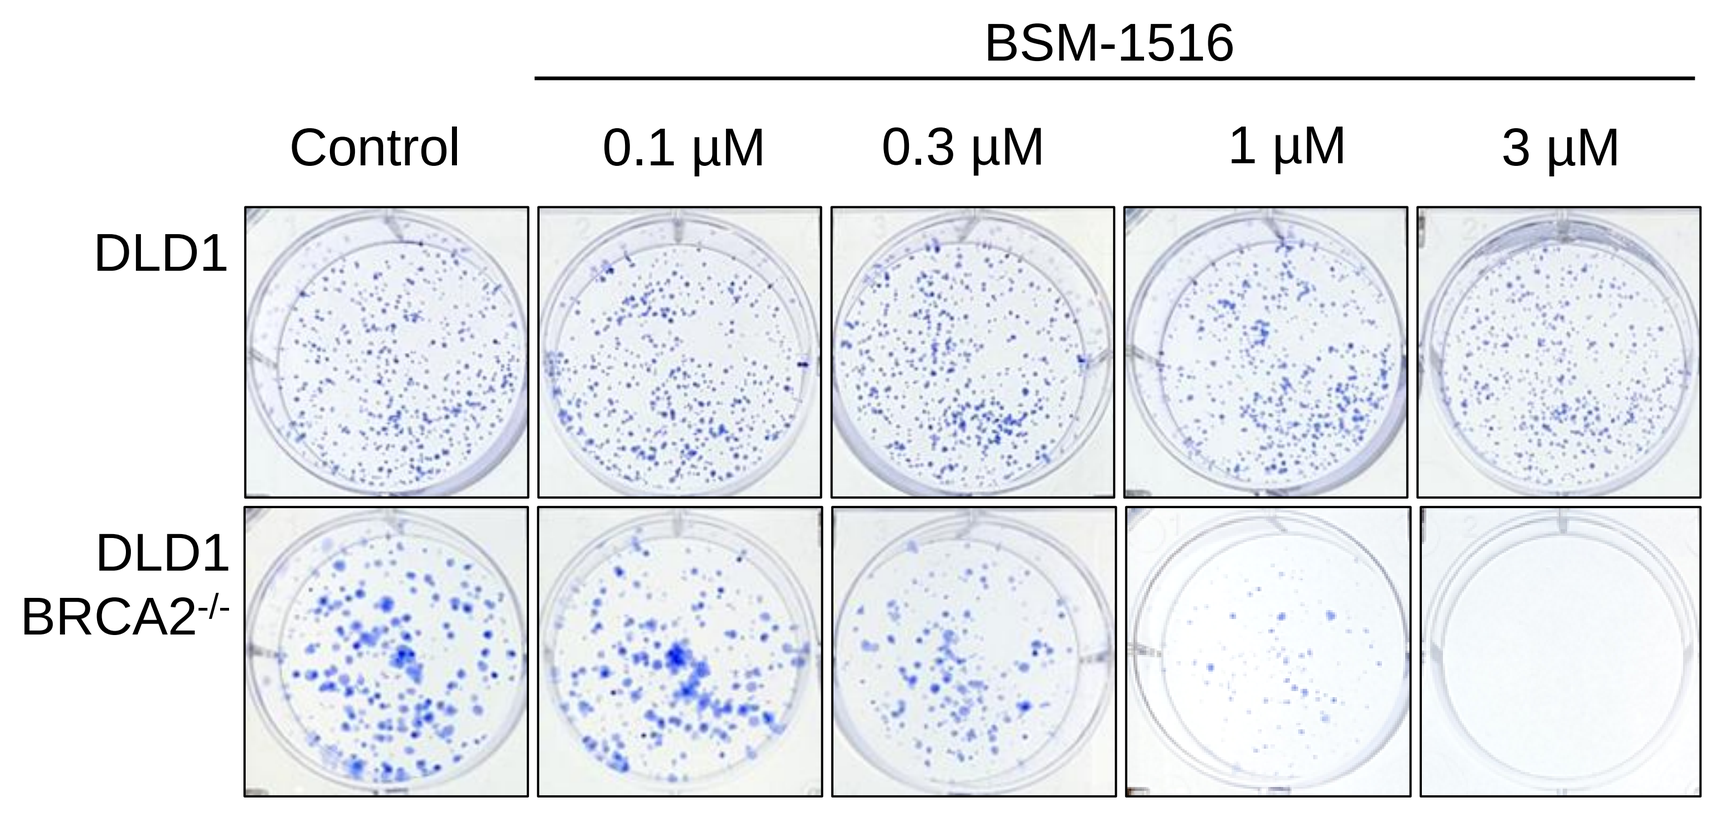
BSM-1516
Control
0.1 µM
0.3 µM
1 µM
3 µM
DLD1
DLD1 BRCA2-/-

Clonogenic survival assay in cell lines treated for 6 days (DLD1) and 12 days (DLD1 $BRCA2^{-/-}$) performed in six-well plates with media change every 3 days visualized and quantified by crystal violet staining.

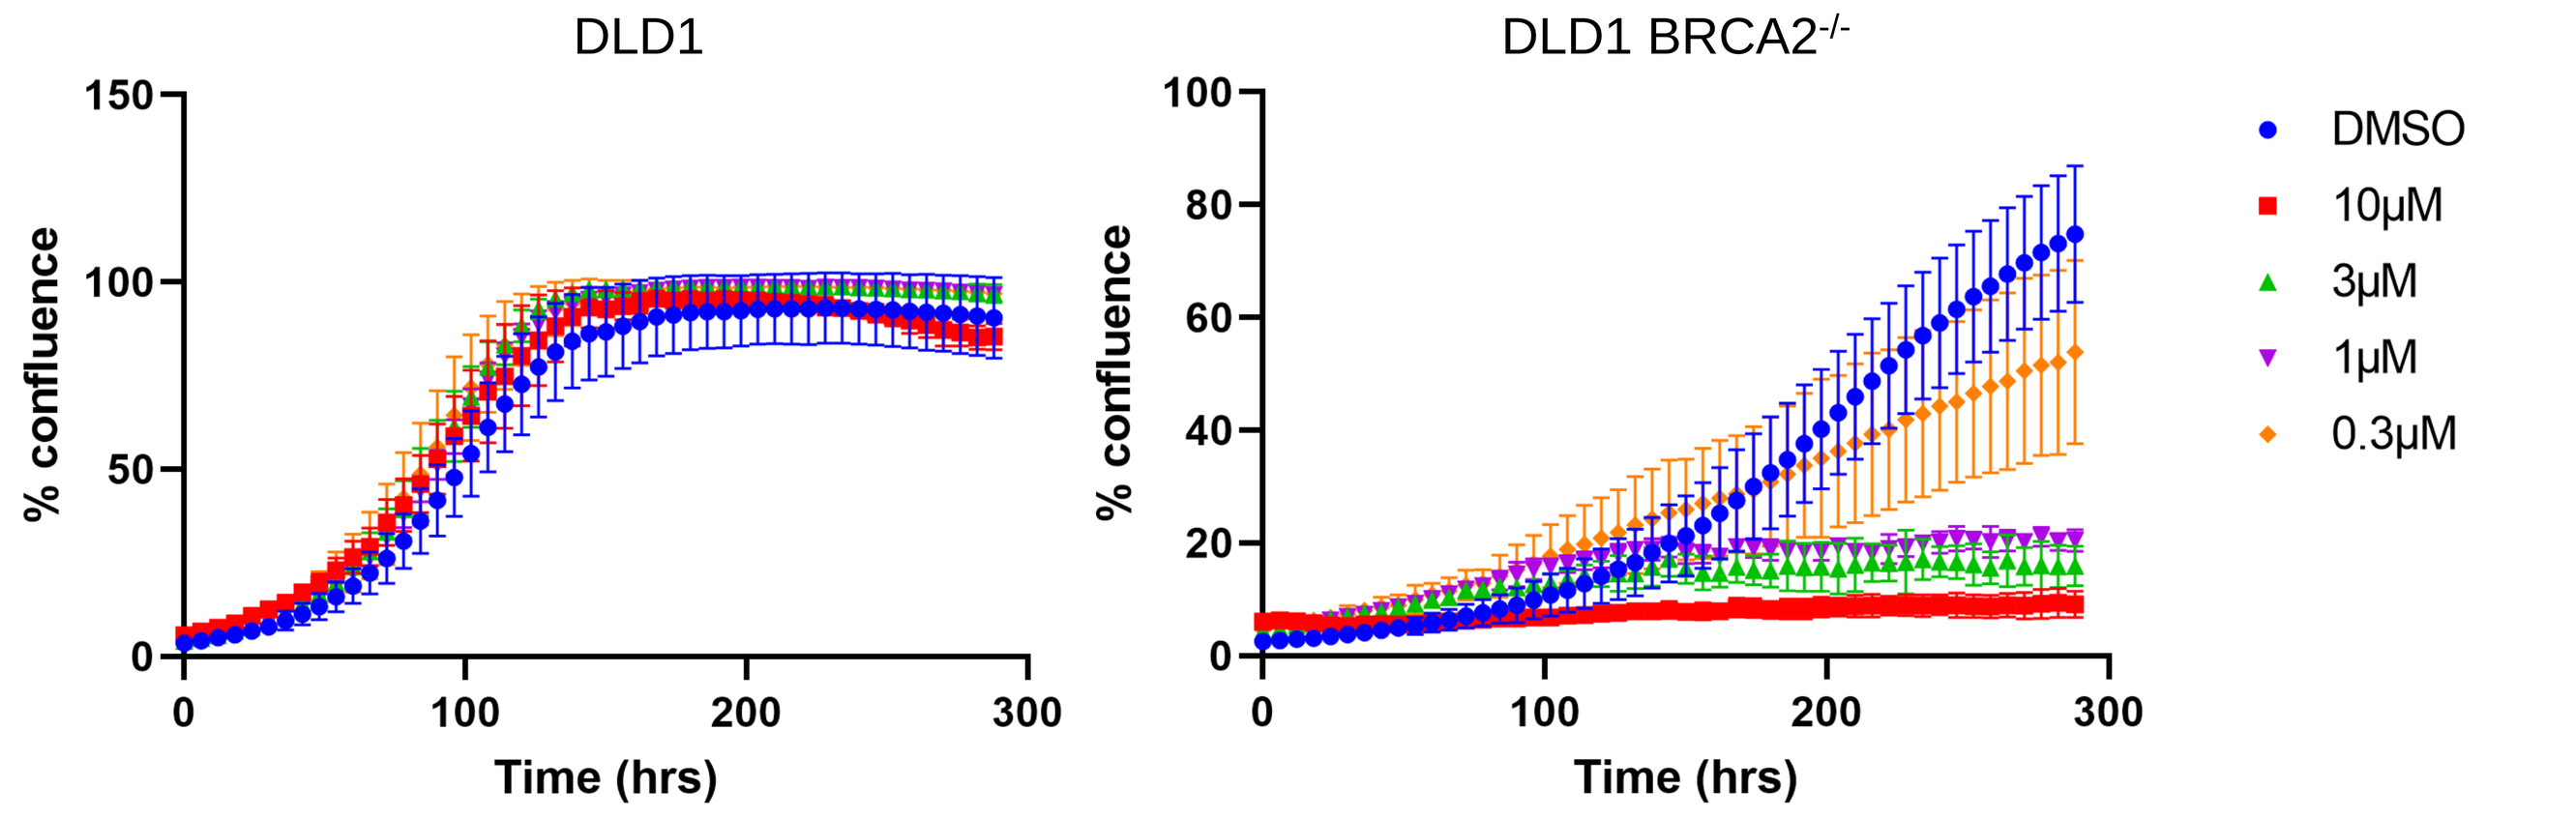
DLD1
DLD1 BRCA2-/-
% confluence
Time (hrs)
DMSO
10µM
3µM
1µM
0.3µM

Real-time cell proliferation growth curves were generated by confluence imaging every 6 hours with triplicate replicates on Incucyte live-cell analysis system.

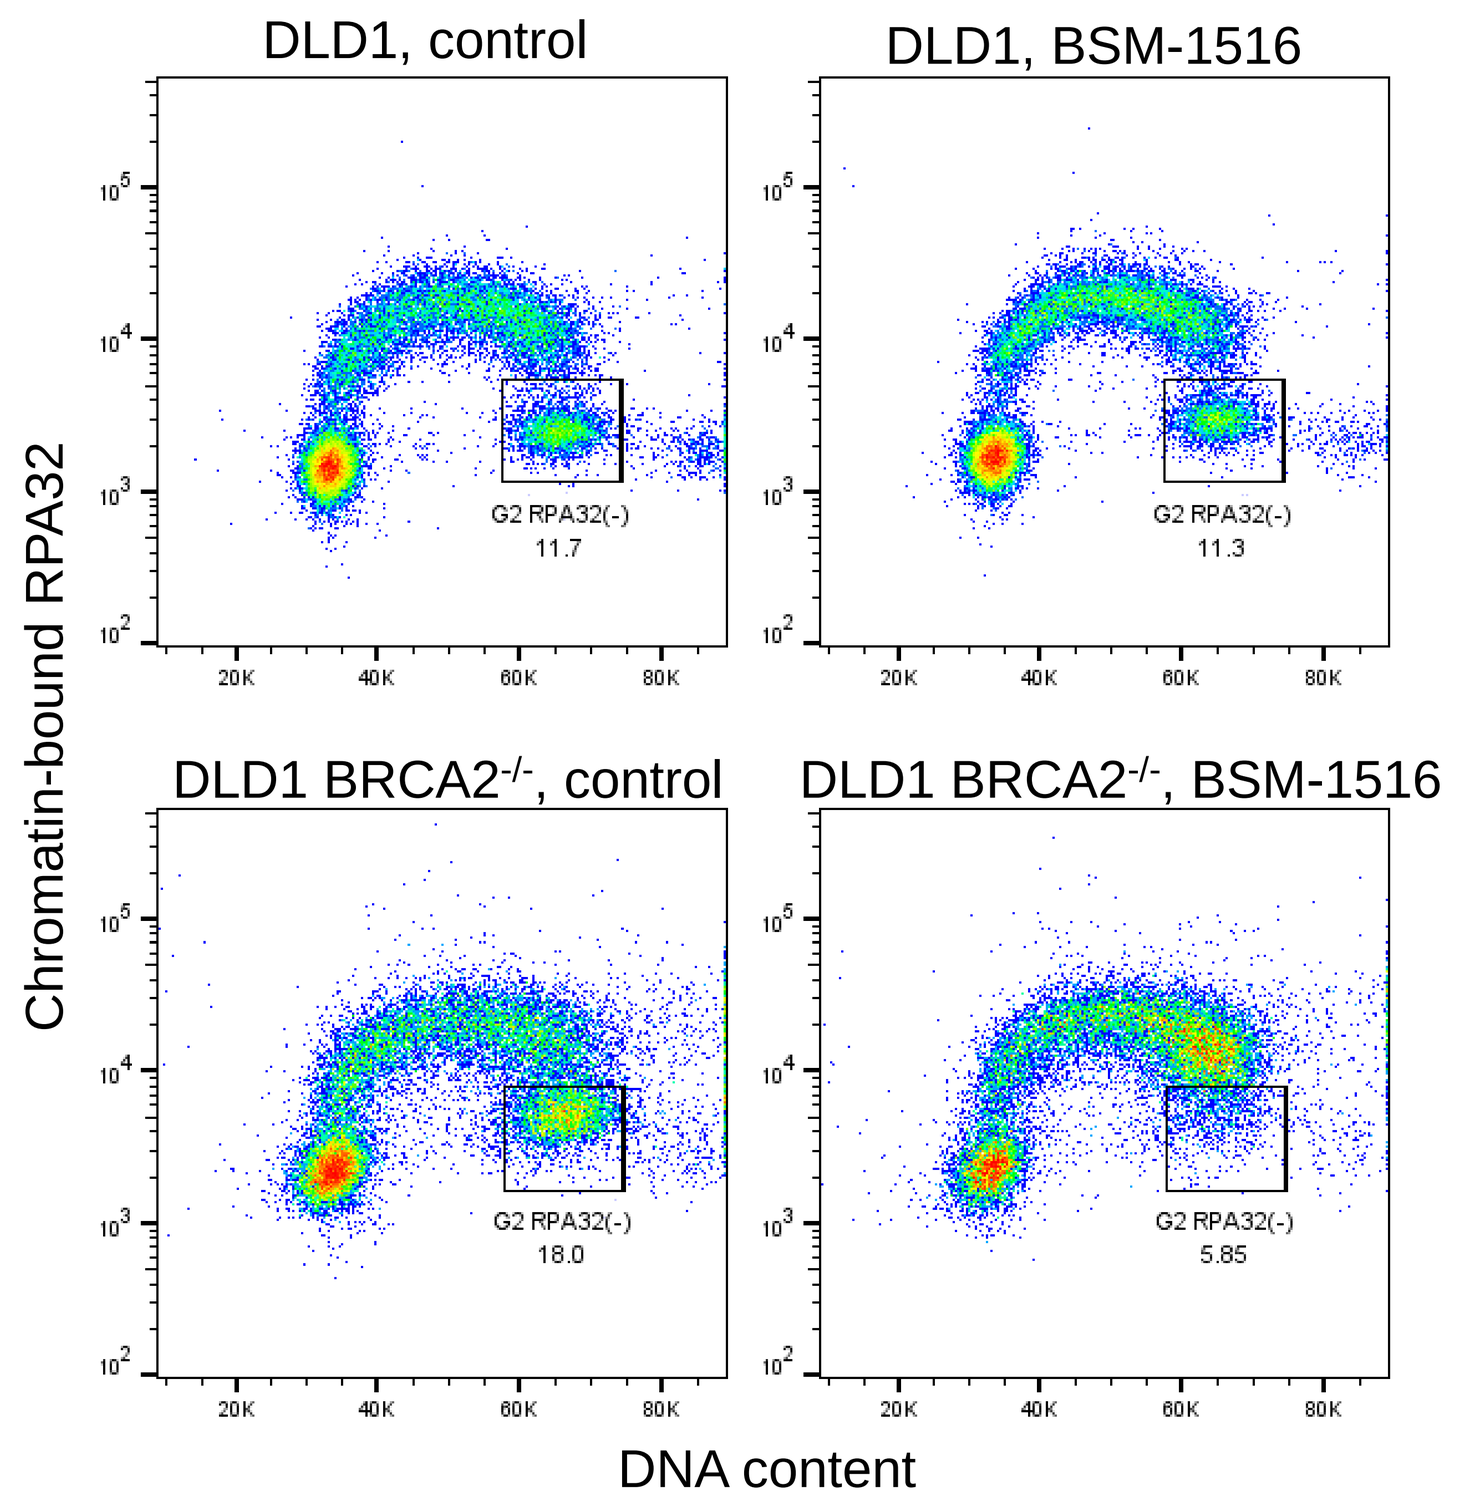
DLD1, control
DLD1, BSM-1516
DLD1 BRCA2-/-, control
DLD1 BRCA2-/-, BSM-1516
G2 RPA32(-) 11.7
G2 RPA32(-) 11.3
G2 RPA32(-) 18.0
G2 RPA32(-) 5.85
Chromatin-bound RPA32
DNA content

Intracellular staining of chromatin-bound RPA32 in DLD1 and DLD1 $BRCA2^{-/-}$ cells treated with 3µM BSM-1516 for 16h. Soluble RPA32 was extracted with detergent prior to fixation and subsequent analysis by flow cytometry.

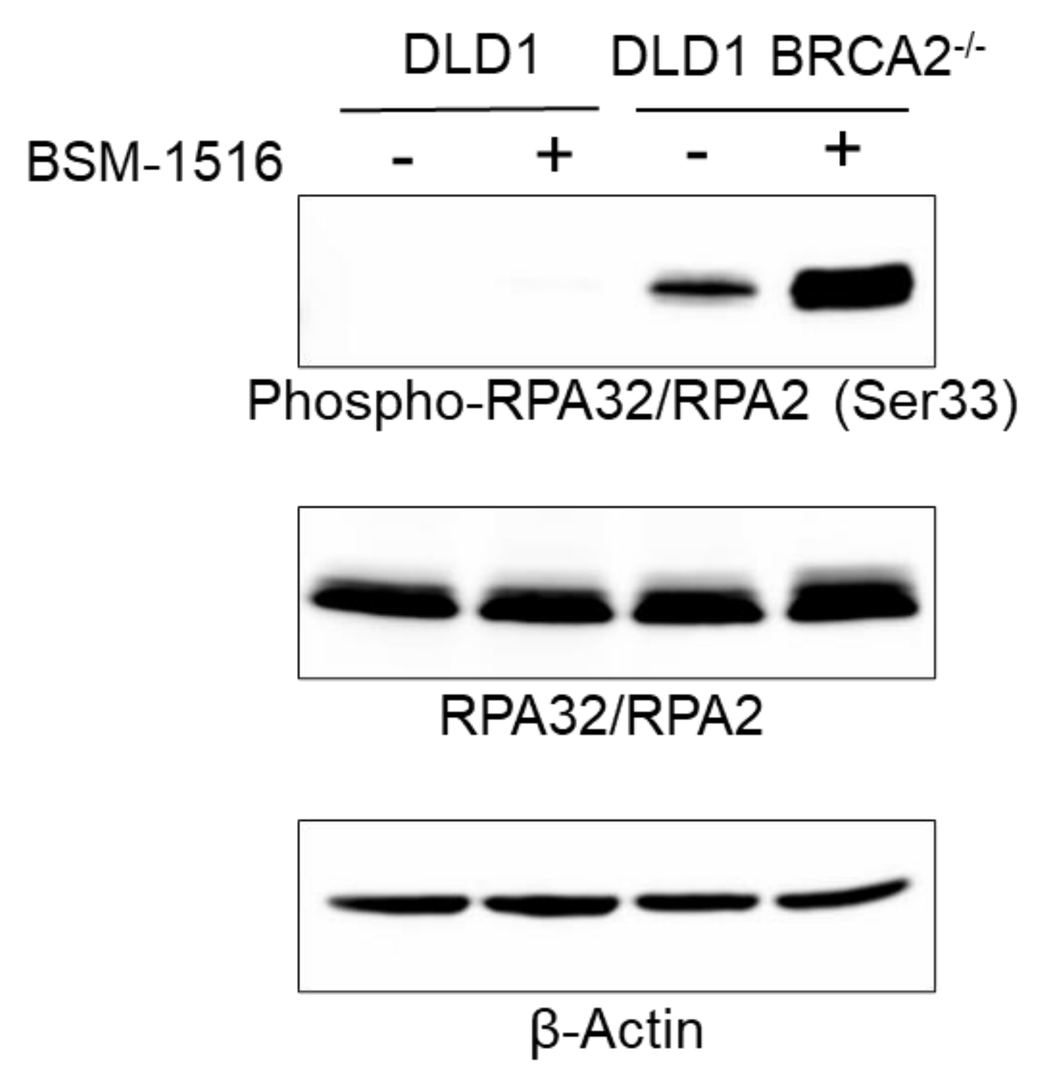
DLD1
DLD1 BRCA2-/-
BSM-1516
Phospho-RPA32/RPA2 (Ser33)
RPA32/RPA2
β-Actin

Western Blot analysis of Ser33-phosphorylated RPA32, an ATR-mediated modification, in DLD1 and DLD1 $BRCA2^{-/-}$ cells treated with 3µM BSM-1516 for 24h.

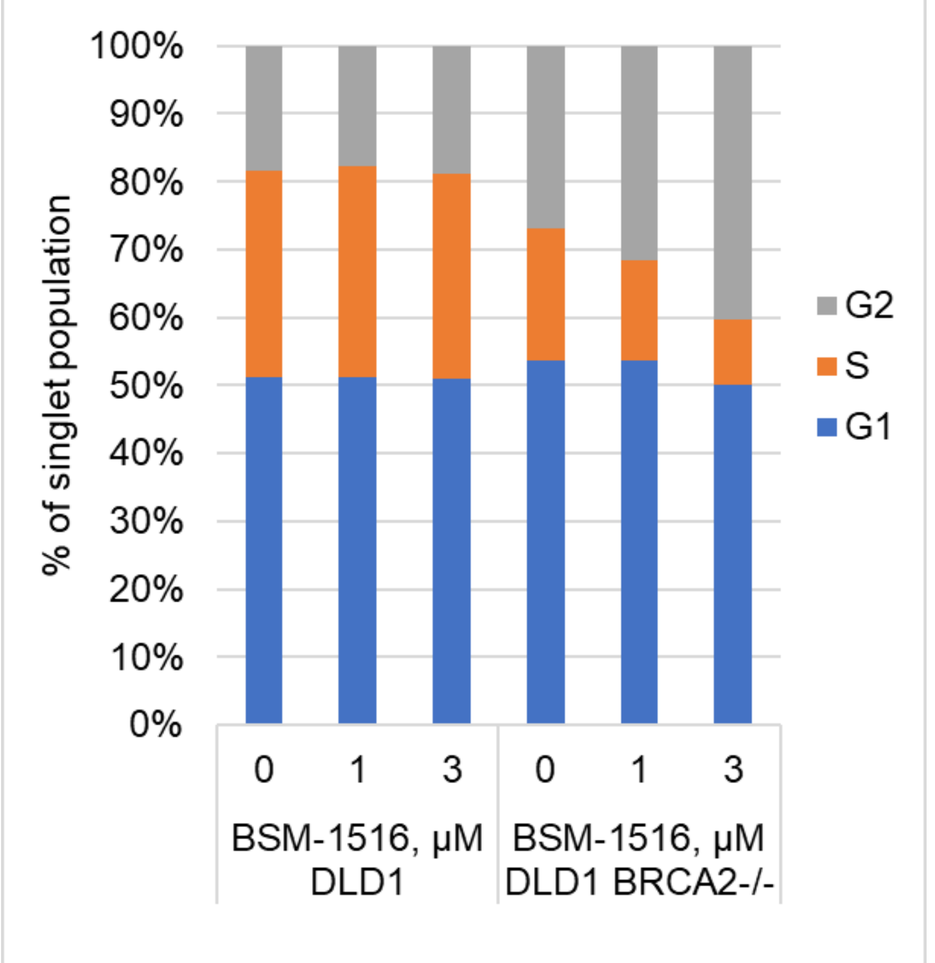
% of singlet population
G2
S
G1
BSM-1516, µM DLD1
BSM-1516, µM DLD1 BRCA2-/-

Pharmacological inhibition of FEN1 by BSM-1516 resulted in G2/M cell cycle arrest of BRCA2–deficient DLD1 cells. Analysis of BrdU incorporation and total DNA content was monitored by flow-cytometry.

Drug combination data: BSM-1516 synergizes with inhibitors of USP1, PARP, PARG, ATR and Chk1

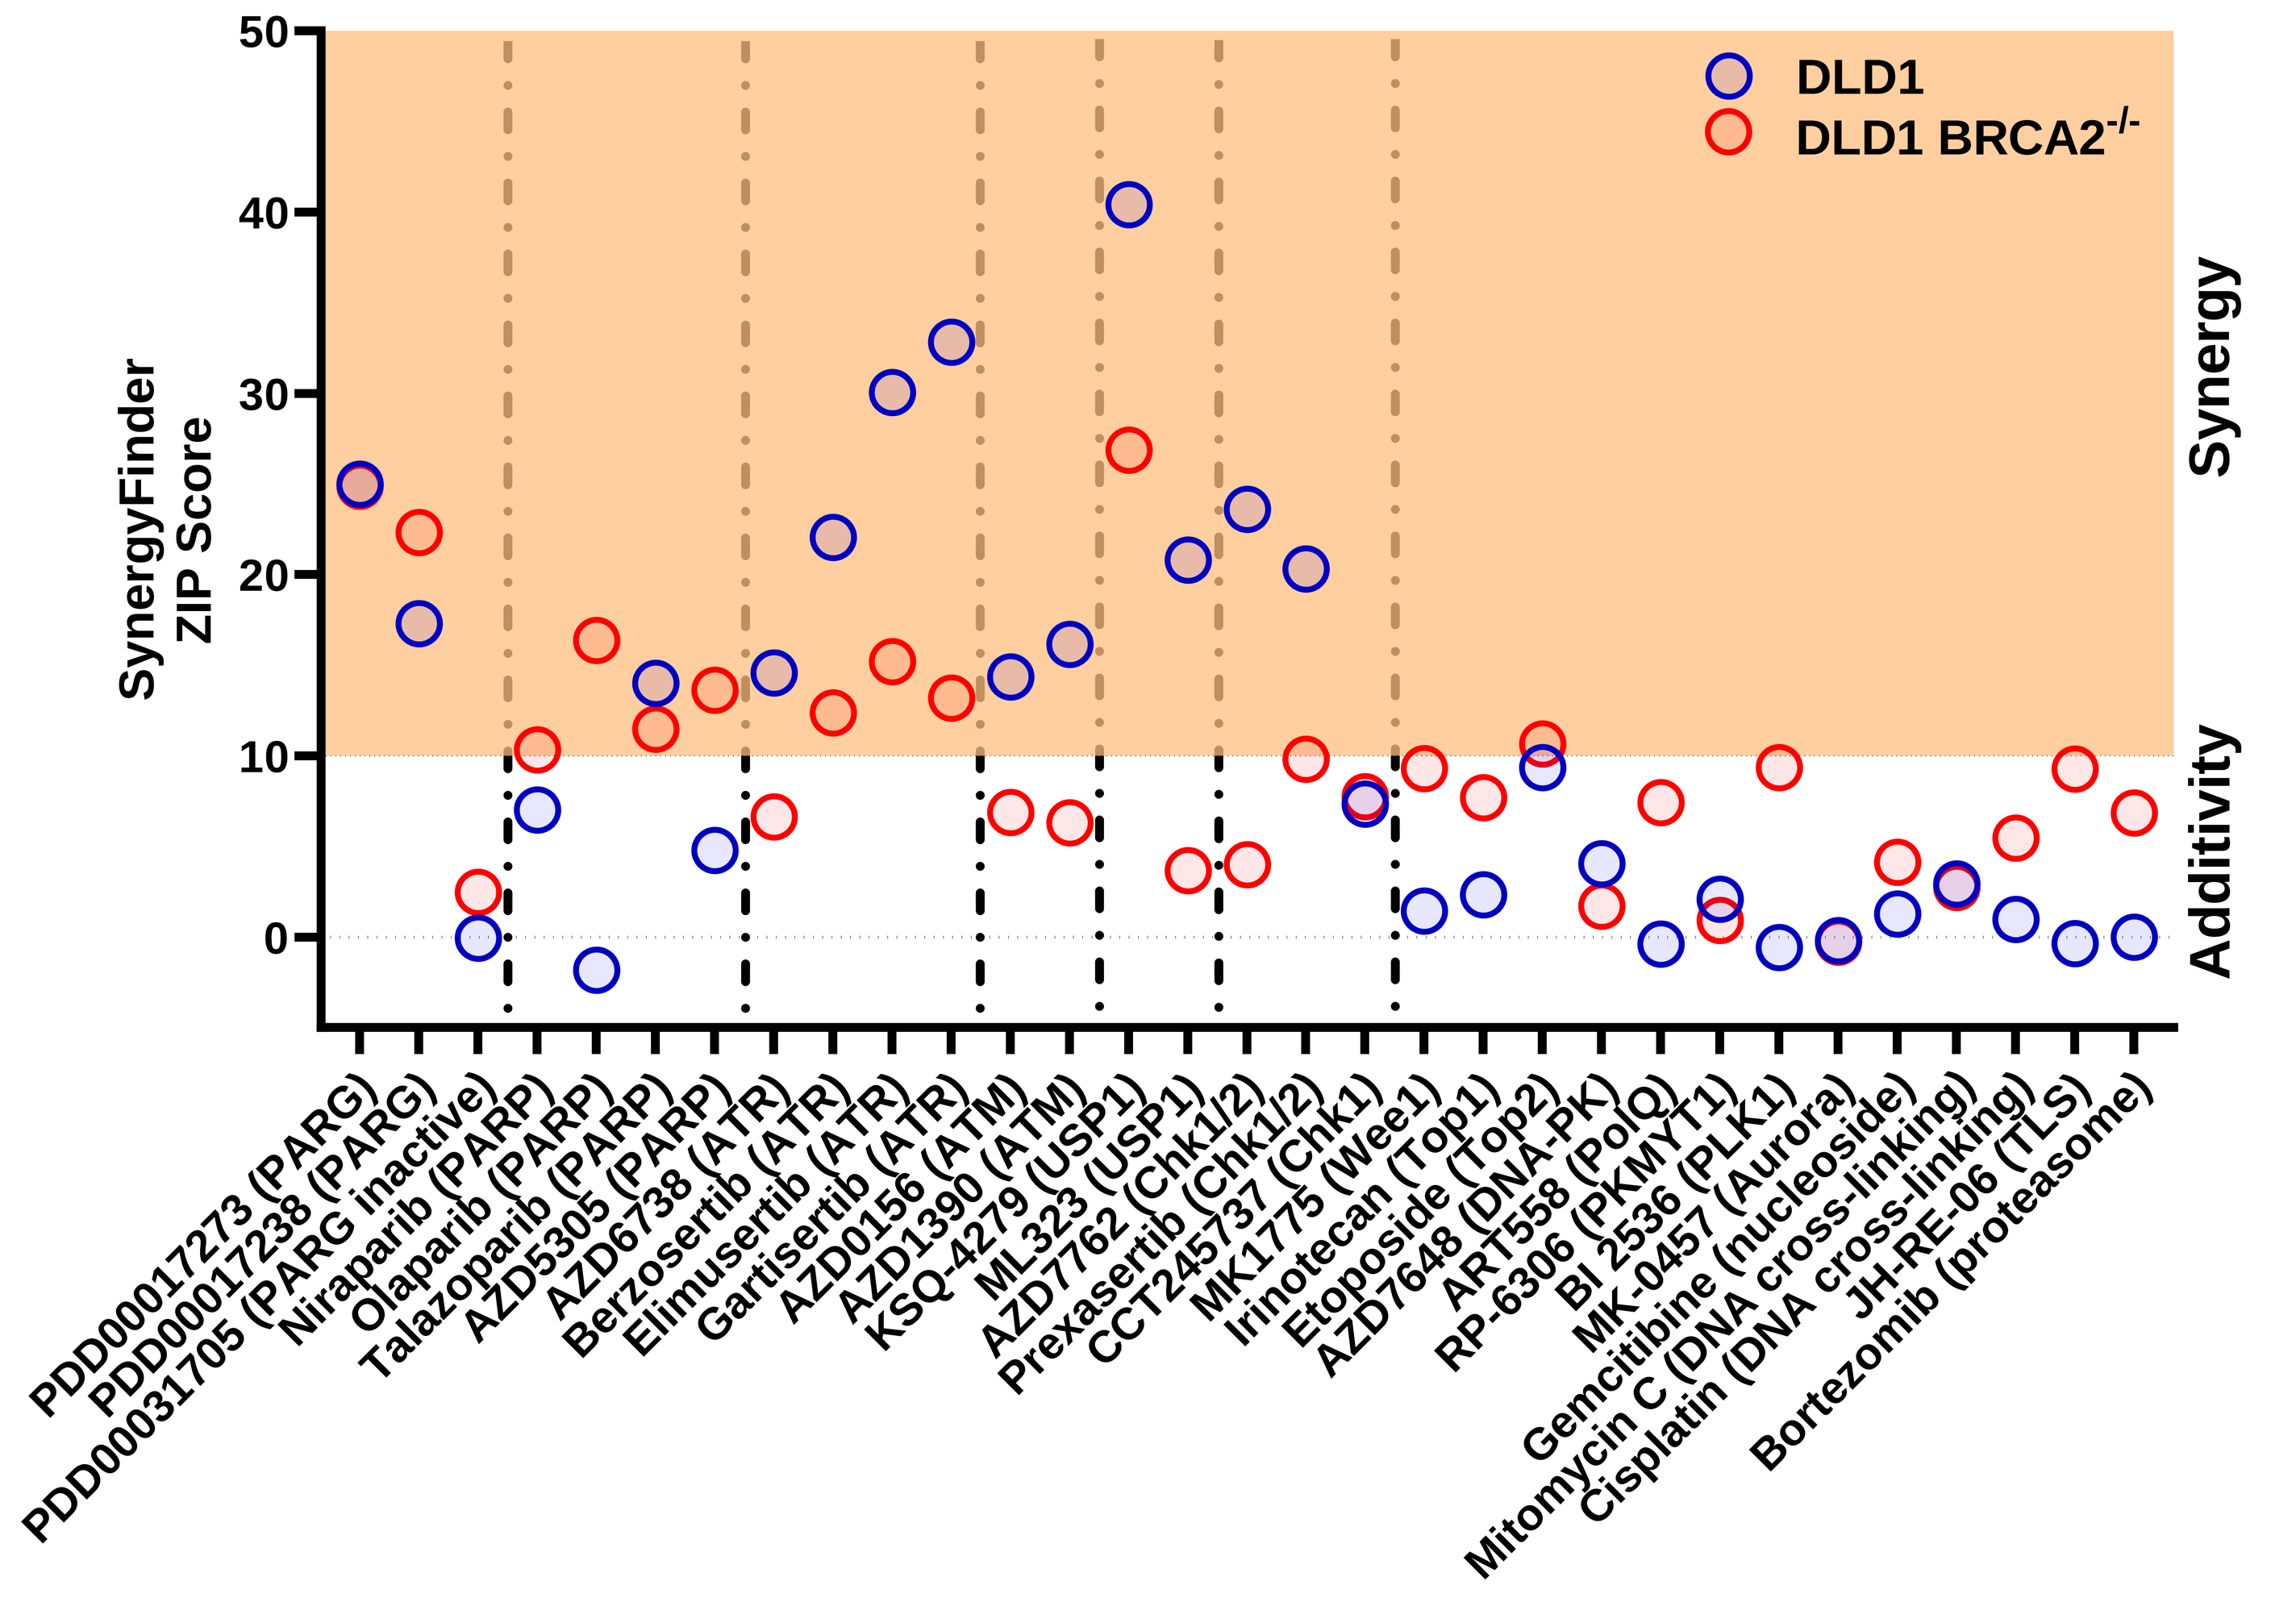
DLD1
DLD1 BRCA2-/-
SynergyFinder ZIP Score
Synergy
Additivity
PDD00017273 (PARG)
PDD00017238 (PARG)
PDD00031705 (PARG inactive)
Niraparib (PARP)
Olaparib (PARP)
Talazoparib (PARP)
AZD5305 (PARP)
AZD6738 (ATR)
Berzosertib (ATR)
Elimusertib (ATR)
Gartisertib (ATR)
AZD0156 (ATM)
AZD1390 (ATM)
KSQ-4279 (USP1)
ML323 (USP1)
AZD7762 (Chk1/2)
Prexasertib (Chk1/2)
CCT245737 (Chk1)
MK1775 (Wee1)
Irinotecan (Top1)
Etoposide (Top2)
AZD7648 (DNA-PK)
ART558 (PolQ)
RP-6306 (PKMYT1)
BI 2536 (PLK1)
MK-0457 (Aurora)
Gemcitibine (nucleoside)
Mitomycin C (DNA cross-linking)
Cisplatin (DNA cross-linking)
JH-RE-06 (TLS)
Bortezomib (proteasome)

To explore BSM-1516 combination potential we performed combination treatment screen with a panel of DNA damage inhibitors and agents. Cell lines were assayed in 6x10 combination half-log dose-response matrix using CellTiter-Glo® 2.0 Cell Viability Assay for 6 days (DLD1) and 9 days (DLD1 $BRCA2^{-/-}$) with media change every 3 days. Synergy scores were calculated using Synergy Finder.

Summary

- Blacksmith Medicines FBDD approach for metalloenzymes identified a series of cell active FEN1-selective inhibitors that use a novel metal-binding pharmacophore.
- Pharmacologic inhibition of FEN1 is synthetic lethal with defects in HRR and synergizes with inhibitors of USP1, PARP, PARG, ATR and Chk1.
- BSM-1516 exhibits favorable *in vivo* PK properties and its in vivo efficacy testing is underway.

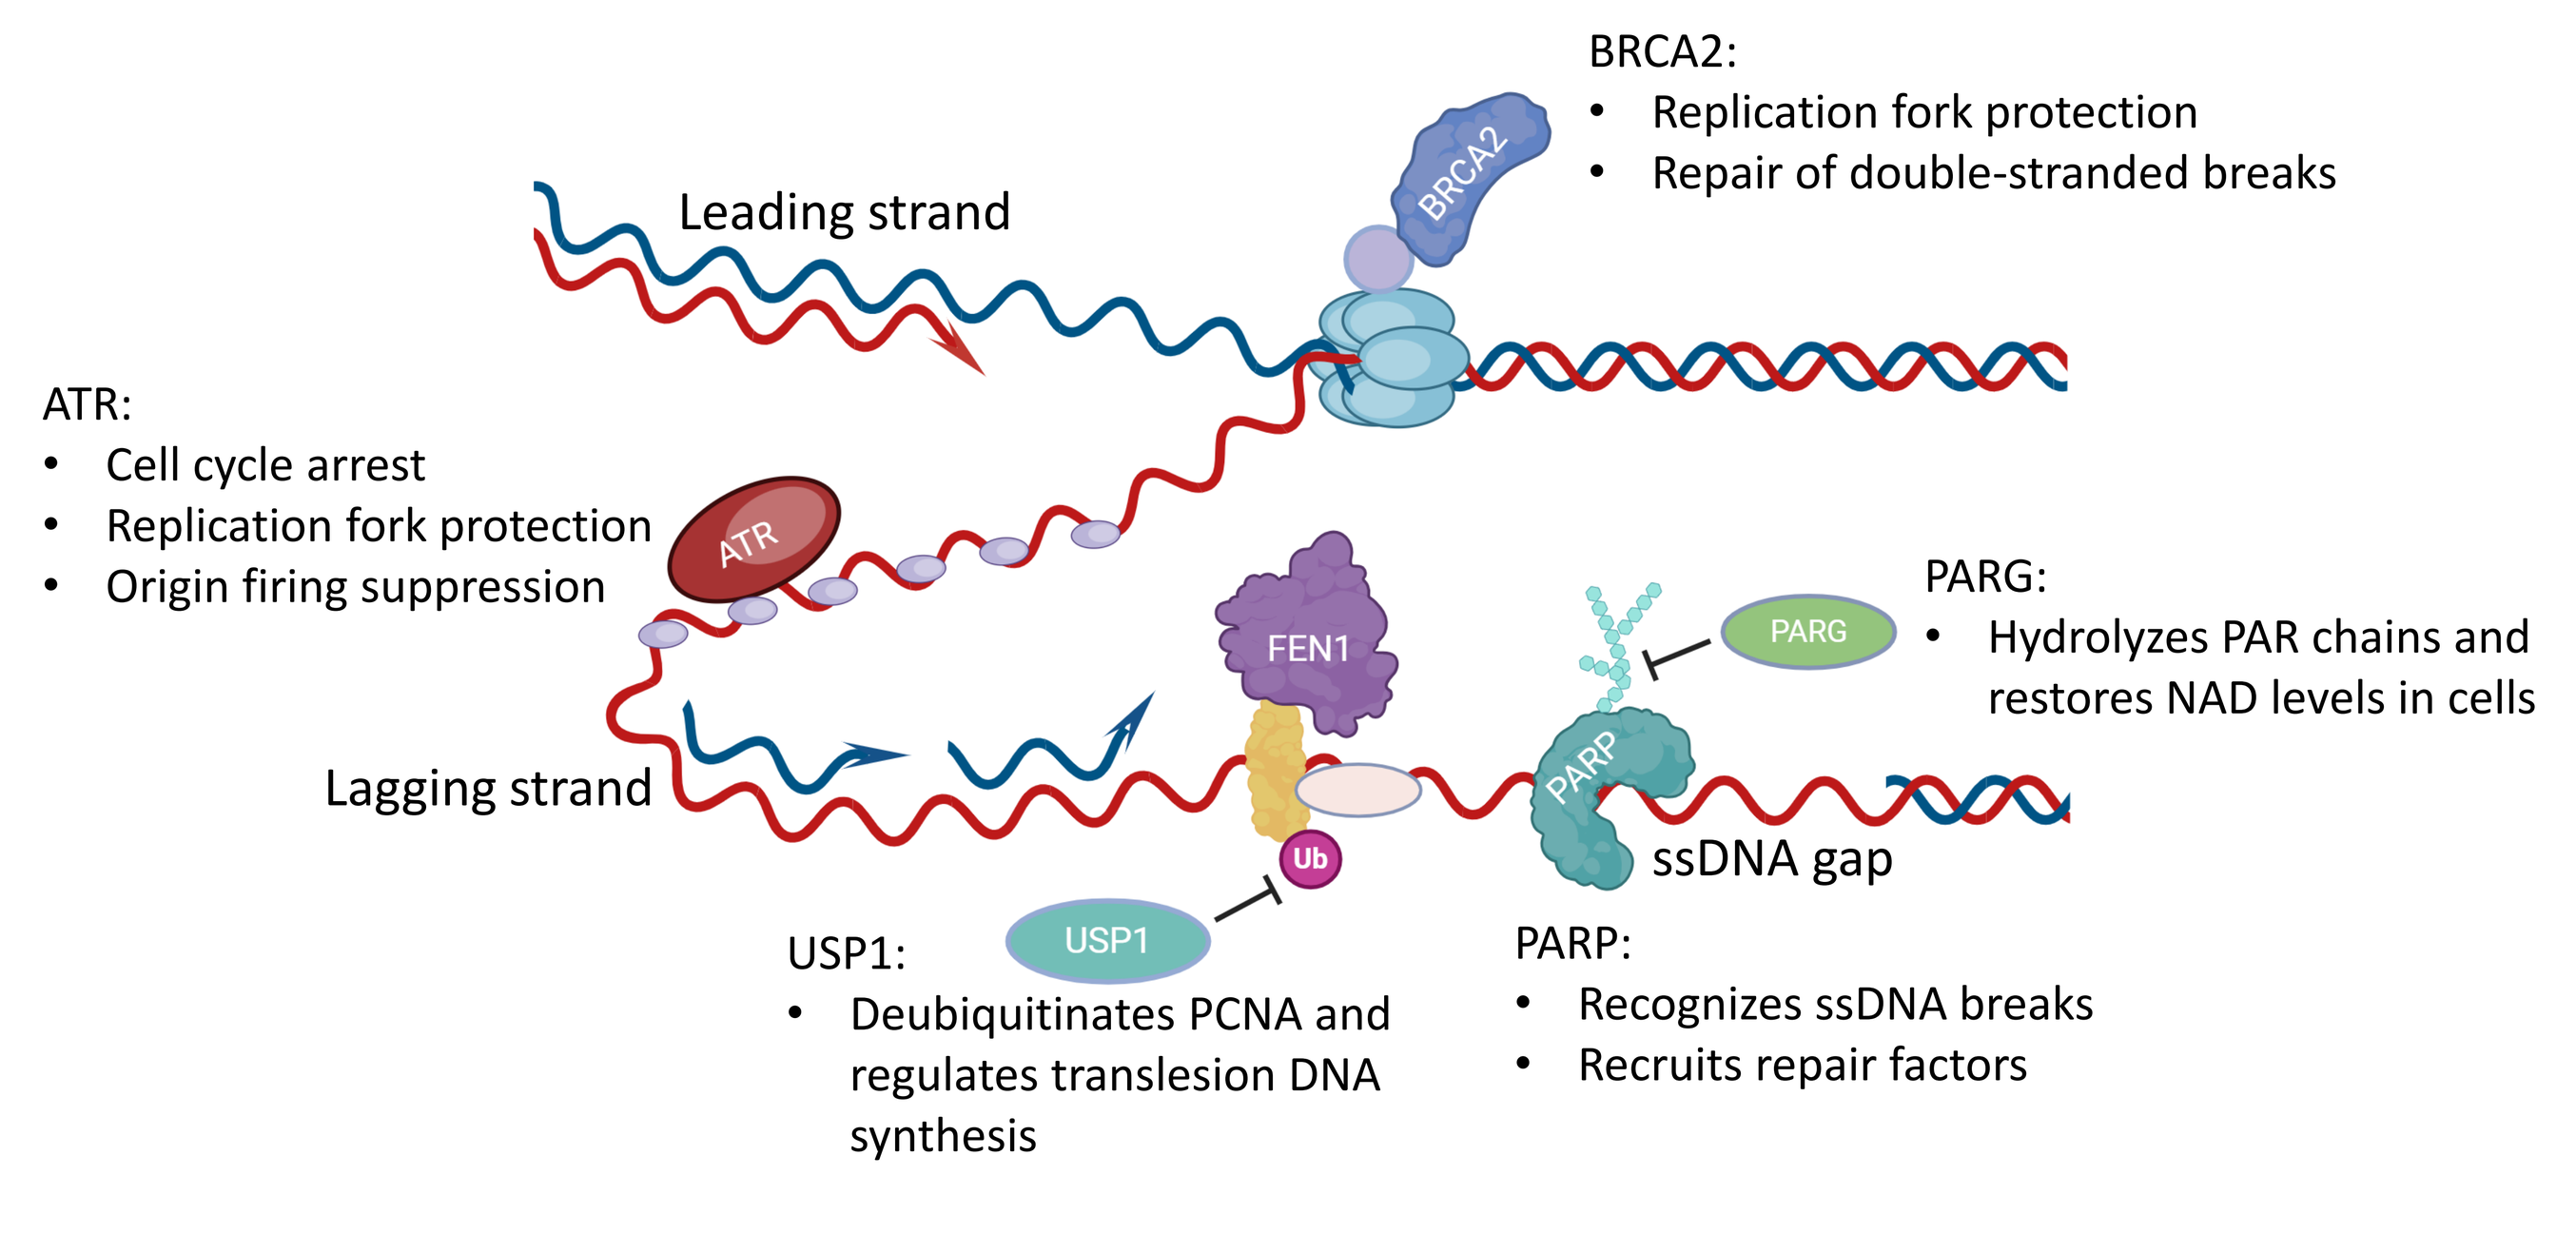
BRCA2:
Replication fork protection
Repair of double-stranded breaks
Leading strand
ATR:
Cell cycle arrest
Replication fork protection
Origin firing suppression
PARG:
Hydrolyzes PAR chains and restores NAD levels in cells
Lagging strand
ssDNA gap
USP1:
Deubiquitinates PCNA and regulates translesion DNA synthesis
PARP:
Recognizes ssDNA breaks
Recruits repair factors

Acknowledgements

Authors are grateful to Chris Lord, Jeff Hager, Neil Gibson, Mark Whittaker, Seth Cohen, Stephen Durant / Evotec DDR team, James Karras, Min Teng, Baskar Nammalwar for support and helpful discussions.